HuOHX10 MultYbody™, an Fc-Engineered Hexameric Anti-OX40 IgG Antibody, Is a Potent Agonist for T Cell Activation

Naoya Tsurushita, Duy Pham, Melinda Sinata, Minami K. Yamamura, and J Yun Tso

JN Biosciences, Mountain View, California

Abstract

JN Biosciences has developed a proprietary Fc engineering technology that allows conversion of a monomeric divalent IgG antibody to a multimeric polyvalent form. Such engineered IgG antibodies (MultYbodies™) can efficiently cross-link TNF receptor super-family members, including costimulatory molecules and death receptors, which require trimerization for induction of intracellular signal transduction. We have applied this technology to generate a hexameric humanized anti-OX40 IgG antibody (HuOHX10 MultYbody™). HuOHX10 MultYbody™, which is composed of Fc-modified heavy and intact light chains, was expressed in CHO-K1 cells and purified from culture supernatants by protein A affinity chromatography. HuOHX10 MultYbody™ enhanced IL-2 expression in human T cells treated with anti-CD3 and anti-CD28 antibodies. In contrast, the IgG1 form of the anti-OX40 antibody had little effects on IL-2 expression. In addition, a synergistic effect was observed between HuOHX10 MultYbody™ and an anti-PD-1 antagonist IgG antibody for IL-2 expression in human PBMC. HuOHX10 MultYbody™, alone or together with a checkpoint blockade antibody, will work as a strong agonist to stimulate immune responses in humans for immunotherapy of cancer and infectious disease. Application of the MultYbody™ technology to generate potent agonist IgG antibodies against other costimulatory molecules, such as CD40, GITR and 4-1BB, is also in progress.

Introduction

The antigen-specific immune response is a complex biological process that is negatively regulated by checkpoint molecules, such as PD-1 and CTLA-4, and positively regulated by costimulatory molecules, such as OX40, GITR and 4-1BB that belong to the TNF receptor super-family (1). Blocking the function of PD-1 or CTLA-4 by an antagonistic monoclonal IgG antibody has been shown to be effective for immunotherapy of cancer in humans (2). TNF receptor super-family members have a unique characteristics that receptor trimerization is required to induce intracellular signal transduction (3), which a conventional divalent IgG antibody cannot achieve by itself. Although Fcγ receptor-mediated cross-linking triggers signal transduction through antibody-bound TNF receptor super-family members, it is not efficient in the body due to the presence of an excess amount of IgG molecules that compete for binding to Fcγ receptors. To overcome this problem for engagement of costimulatory molecules, we have developed proprietary Fc engineering technologies for conversion of IgG antibodies into three different multimeric polyvalent forms (MultYbodies™) that can efficiently cross-link TNF receptor super-family members without the aid of Fcγ receptors for induction of intracellular signal transduction. Using one of these proprietary technologies, we have generated a potent anti-OX40 agonist antibody.

Materials and Methods

Generation of HuOHX10 MultYbody™. A mouse monoclonal antibody OHX10 that binds to human OX40 was generated following standard hybridoma techniques. The VH and VL regions of OHX10 were humanized as previously described (4). The synthetic humanized OHX10 (HuOHX10) VH and VL genes were cloned into two different vectors, one for expression in the human IgG1/kappa form and another in the MultYbody™ form (Fig. 1). In the MultYbody™ expression vector, the CH3 and CH4 regions of the human mu heavy chain (Cμ3 and Cμ4, respectively) are attached to the C-terminal of the human gamma-1 constant region.

Structure of antibody expression vectors

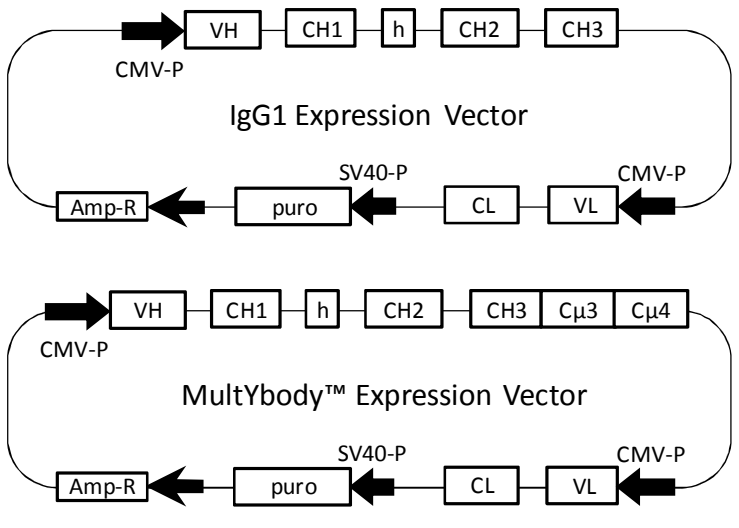
VH
CH1
h
CH2
CH3
CMV-P
IgG1 Expression Vector
SV40-P
CMV-P
Amp-R
puro
CL
VL
Cμ3
Cμ4
MultYbody™ Expression Vector

Figure 1. Schematic structures of IgG1 (top) and MultYbody™ (bottom) expression vectors are shown. Symbols used: CMV-P, CMV promoter; VH, heavy chain variable region; CH1, h, CH2 and CH3, human gamma-1 constant regions; Cμ3 and Cμ4, human Cμ3 and Cμ4 constant regions; VL, light chain variable region; CL, human kappa constant region; SV40-P, SV40 early promoter; puro, puromycin N-acetyl-transferase; Amp-R; beta lactamase.

Materials and Methods (*continued*)

Structure of HuOHX10 MultYbody™

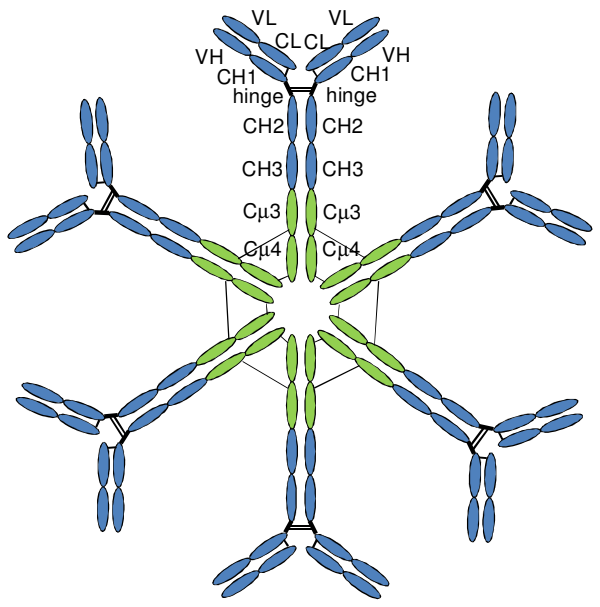
VL
VL
CL CL
VH
VH
CH1
CH1
hinge
hinge
CH2
CH2
CH3
CH3
Cμ3
Cμ3
Cμ4
Cμ4

Figure 2. A schematic structure of HuOHX10 MultYbody™ is shown. The CH3 and CH4 regions of the human mu heavy chain (Cμ3 and Cμ4, respectively) are attached to the C-terminal of the human gamma-1 constant region for conversion to a hexamer. The lines between antibody monomers represent disulfide bonding. Other symbols are described in the legend to Fig. 1.

Expression and purification of HuOHX10 MultYbody™. Each of the expression vectors for HuOHX10 IgG1 and MultYbody™ was stably transfected into CHO-K1 cells. Antibody expression level in culture supernatants was analyzed by ELISA. CHO-K1 stable transfectants were expanded in protein-free media. HuOHX10 IgG1 and MultYbody™ were purified from culture supernatants by protein A affinity chromatography and dialyzed against PBS for storage.

Results

Size analysis of HuOHX10 MultYbody™

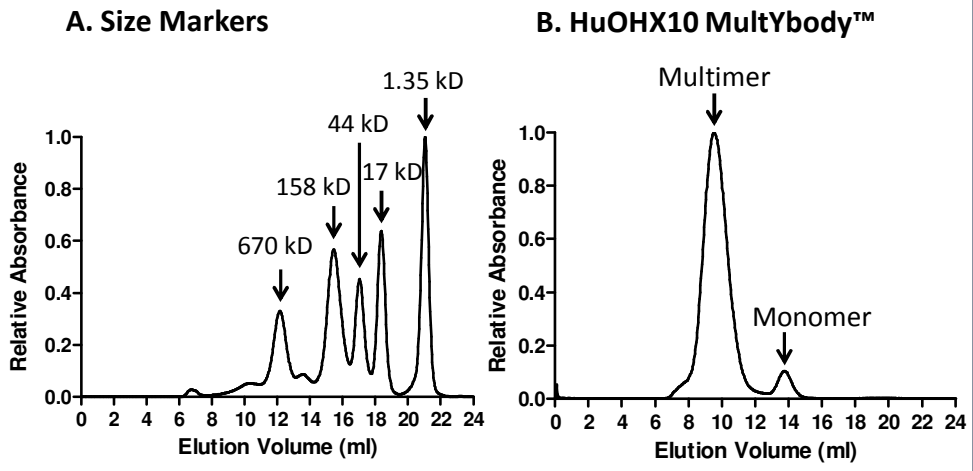
A. Size Markers
1.35 kD
44 kD
158 kD
17 kD
670 kD
Relative Absorbance
Elution Volume (ml)
B. HuOHX10 MultYbody™
Multimer
Monomer
Relative Absorbance
Elution Volume (ml)

Figure 3. The molecular size of protein A-purified HuOHX10 MultYbody™ in the native form was analyzed by gel filtration using the AKTA FPLC system with a Superose 6 10/300 GL column (GE Healthcare, Indianapolis, IN). PBS (pH 7.4) was used as elution buffer. Samples used: Panel A, Gel Filtration Standard (BioRad, Hercules, CA); Panel B, HuOHX10 MultYbody™.

Enhancement of IL-2 expression in CD4+ T cells by HuOHX10 MultYbody™

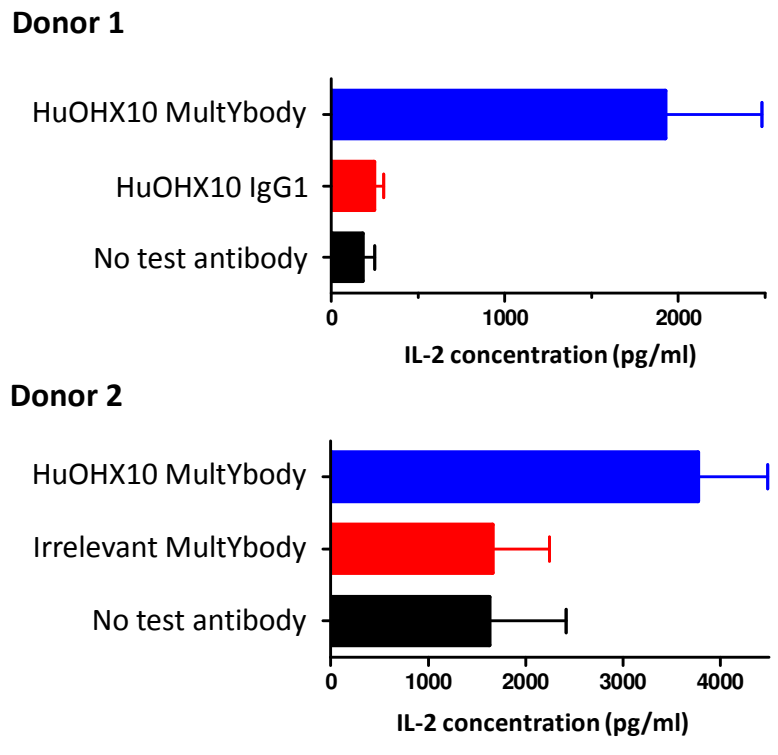
Donor 1
HuOHX10 MultYbody
HuOHX10 IgG1
No test antibody
0
1000
2000
IL-2 concentration (pg/ml)
Donor 2
HuOHX10 MultYbody
Irrelevant MultYbody
No test antibody
0
1000
2000
3000
4000
IL-2 concentration (pg/ml)

Figure 4. Human PBMC-derived CD4+ T cells were treated with 1 μg/ml each of anti-CD3 (plate-coated) and anti-CD28 (in media) monoclonal antibodies in the presence (or absence) of 1 μg/ml of a test antibody in media. IL-2 levels in culture supernatants were measured by ELISA after 2-day incubation with a test antibody.

Results (*continued*)

Synergy with an anti-PD-1 antagonist antibody for IL-2 expression in PBMC

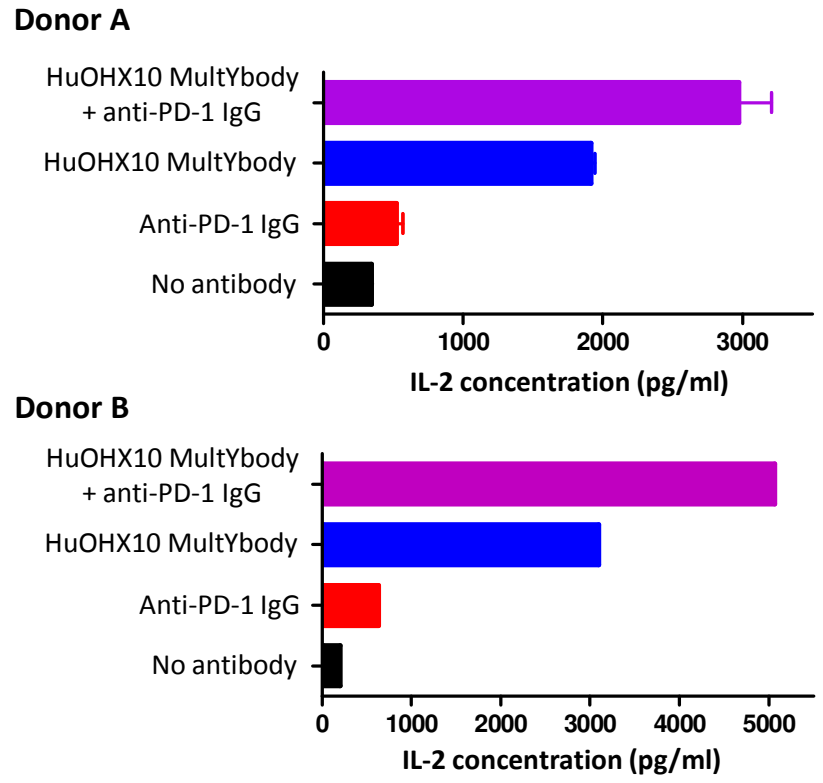
Donor A
HuOHX10 MultYbody
+ anti-PD-1 IgG
HuOHX10 MultYbody
Anti-PD-1 IgG
No antibody
0
1000
2000
3000
IL-2 concentration (pg/ml)
Donor B
HuOHX10 MultYbody
+ anti-PD-1 IgG
HuOHX10 MultYbody
Anti-PD-1 IgG
No antibody
0
1000
2000
3000
4000
5000
IL-2 concentration (pg/ml)

Figure 5. Human PBMC were treated with 1 μg/ml each of anti-CD3 (plate-coated for Donor A, in media for Donor B) and anti-CD28 (in media) monoclonal antibodies in the presence of (i) 200 ng/ml of HuOHX10 MultYbody™, (ii) 1 μg/ml of a mouse anti-PD-1 antagonist IgG antibody, (iii) 200 ng/ml of HuOHX10 MultYbody™ and 1 μg/ml of a mouse anti-PD-1 antagonist IgG antibody, and (iv) no antibody in media. IL-2 levels in culture supernatants were measured by ELISA after 2-day incubation with the test antibody.

Conclusion

- A recombinant hexameric humanized anti-OX40 IgG antibody (HuOHX10 MultYbody™), which can efficiently cross-link OX40 molecules on the cell surface without the aid of Fcγ receptors, was generated using JN Biosciences' proprietary Fc engineering technology (Figs. 1, 2 and 3).
- HuOHX10 MultYbody™ enhanced IL-2 expression in human CD4+ T cells treated with anti-CD3 and anti-CD28 antibodies, while the IgG form of the anti-OX40 antibody showed little effects on IL-2 expression (Fig. 4).
- HuOHX10 MultYbody™ enhanced IL-2 expression synergistically with an anti-PD-1 antagonist antibody in human PBMC (Fig. 5).
- The Fc engineering technology used in this work is applicable to generation of agonist antibodies against other costimulatory molecules for immunotherapy of cancer and infectious disease.

Highlights of MultYbody™

- Composed of Fc-modified heavy and intact light chains with no additional polypeptides
- Expressed in mammalian cell stable transfectants and purified from culture supernatants by protein A
- Requires no modifications after protein A purification
- Potently induces signal transduction by multivalent cross-linking of TNF receptor super-family members
 - Induction of apoptosis by MultYbodies™ against death receptors such as DR4 and DR5
 - Enhancement of T cell activation by MultYbodies™ against costimulatory molecules such as OX40, GITR and 4-1BB
 - Enhancement of the activation of antigen-presenting cells by MultYbodies™ against CD40
- A humanized anti-DR4 MultYbody was more efficacious than the IgG1 form to prolong mouse survival in xenograft models of lymphoma and myeloma (5)
- Amenable to generation of bispecific antibodies
- A novel class of therapeutic antibodies

References

1. Mahoney et al. Nat. Rev. Drug Discov. 14:561-584 (2015)
2. Shin et al. Curr. Opin. Immunol. 33:23-35 (2015)
3. Croft et al. Nat. Rev. Drug Discov. 12:147-168 (2013)
4. Tsurushita et al. Methods 36:69-83 (2005)
5. http://www.jn-bio/pipeline/huyon007

Contact Information

Naoya Tsurushita, Ph.D.
JN Biosciences LLC
320 Logue Avenue, Mountain View, CA 94043, U.S.A.
TEL: 650-967-9486
Email: ntsurushita@jn-bio.com

© JN Biosciences 2015